No Detected Resistance to Tenofovir Alafenamide (TAF) Through 96 Weeks of Treatment in Children and Adolescents With Chronic Hepatitis B

1429-C

Philip Rosenthal[1], Mei-Hwei Chang[2], Jorge Bezerra[3], Yang Liu[4], Dong Han[4], Simin Xu[4], Caleb Marceau[4], Tahmineh Yazdi[4], Lindsey May[4], Robert Li[4], Savrina Manhas[4], Pui Yan Ho[4], Clarissa Martinez[4], Nadine Peinovich[4], Silvia Chang[4], Ross Martin[4], Evguenia Maiorova[4], Hongmei Mo[4], Frida Abramov[4], John F Flaherty[4], Kathleen Schwarz[5]

[1]University of California San Francisco, San Francisco, CA, USA; [2]National Taiwan University and Children Hospital, Taipei, Taiwan; [3]University of Texas Southwestern Medical Center, Dallas, TX, USA; [4]Gilead Sciences, Inc., Foster City, CA, USA; [5]UC San Diego School of Medicine, Rady Children's Hospital–San Diego, San Diego, CA, USA

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

Key Findings

- **Among 88 pediatric patients treated with TAF for 2 years (96 weeks), 24 (27%) qualified for sequence analysis; 12 (50%) qualified due to persistent viremia (HBV DNA ≥69 IU/mL)**
- **Of 24 patients, 17 (71%) showed either no sequence change in pol/RT from baseline or were unable to be sequenced at week 96; conserved site changes were observed in only 3 (13%) patients**
- **Week 96 isolates from the patients who qualified for phenotypic analysis (n = 5) remained sensitive to TAF in vitro (<2-fold change in EC_{50} vs baseline)**

Conclusions

No resistance to TAF was detected through 96 weeks of treatment in adolescents and children with CHB

References: **1.** EASL 2017 Clinical Practice Guidelines on the Management of Hepatitis B Virus Infection. *J Hepatol*. 2017;67:370-98. **2.** Jonas MM, et al. *Hepatology*. 2016;63:377-87. **3.** Murray KF, et al. *Hepatology*. 2012;56:2018-26. **4.** Terrault NA, et al. *Hepatology*. 2018;67:1560-99. **5.** Vemlidy [package insert]. Gilead Sciences, Inc. 2022. Accessed August 25, 2023. **6.** Vemlidy. Summary of product characteristics. European Medicines Agency. Accessed August 25, 2023. **7.** Cathcart AL, et al. *Antimicrob Agents Chemother*. 2018;62(10):e01064-18. **8.** Chan HCY, et al. Poster 1430-C presented at AASLD 2023. **9.** Schwarz K, et al. AASLD 2022.

Acknowledgments: This study was funded by Gilead Sciences, Inc., Foster City, CA. Medical writing and editorial support were provided by Helen Rodgers, PhD, of AlphaScientia, a Red Nucleus company, and were funded by Gilead Sciences, Inc.

We extend our thanks to the patients, their families, and all participating investigators: Belgium: E Sokal; Hong Kong: GLH Wong; India: A Chowdhury, R Mehta, Dr Shalimar, A Shukla, M Tandon; New Zealand: H Evans; Republic of Korea: BH Choe, YH Choe, KM Kim; Romania: D Pacurar, A Streinu-Cercel; Russian Federation: I Khaertynova, Y Lobzin, V Morozov, T Strokova; Taiwan: MH Chang, MW Lai, HH Shih, JF Wu, YJ Yang; USA: J Bezerra, J Daniel, L Gillis, JR Honegger, S Horslen, CH Lin, N Mittal, D Mogul, K Murray, MR Narkewicz, D Pan, Y Pham, G Rao, P Rosenthal, K Schwarz, M Whitworth.

Disclosures:
PR reports research grants from Albireo; Arrowhead; Gilead Sciences, Inc.; Merck; Mirum; Takeda; and Travere; and consultant fees from Albireo; Ambys; Audentes; BioMarin; Dicerna; Encoded; Gilead Sciences, Inc.; MedinCell; Mirum; Takeda; and Travere. **MHC** reports no conflicts of interest. **JB** reports research grants from Gilead Sciences, Inc., and consulting fees from DX4 Liver. **YL**, **DH**, **SX**, **CM**, **TY**, **LM**, **RL**, **SM**, **PYH**, **CM**, **NP**, **SC**, **RM**, **EM**, **HM**, **FA**, and **JFF** are employees of Gilead Sciences, Inc., and may own stock or stock options. **KS** reports research grants from Albireo and Gilead Sciences, Inc., and consulting for Gilead Sciences, Inc.; Mirum; Sarepta; and Up to Date.

Introduction

- Children and adolescents with chronic hepatitis B (CHB) have an increased lifetime risk for clinical complications, including progression to cirrhosis, decompensated liver disease, and liver cancer[1]
- First-line treatment options for children with CHB are generally similar to those for adults; however, there are potential safety and efficacy limitations, including reduced response rates and/or development of resistance in patients with prior lamivudine experience treated with entecavir and bone mineral density reductions (relative to placebo [PBO]) with use of tenofovir disoproxil fumarate[2-4]
- The US Food and Drug Administration recently approved tenofovir alafenamide (TAF) for the treatment of CHB in patients aged 12 and older with compensated liver disease,[5] and the European Medicines Agency recently expanded the indication for children aged 6 and older weighing ≥25 kg based on week 48 findings from a randomized, double-blind, controlled trial (GS-US-320-1092; NCT02932150) in children and adolescents[6]
- No resistance to TAF has been observed during double-blind treatment through 96 weeks[7] and through year 8 of open-label TAF treatment in adults with CHB[8]; likewise, no resistance to TAF was detected in adolescents and children with CHB through 48 weeks of TAF treatment[9]

Objective

- To assess the potential for resistance development at week 96 in a Phase 2 study evaluating the adult dose of TAF treatment (25 mg once daily [QD]) when given to adolescents and children with CHB

Methods

- Randomized (2:1), double-blind, PBO-controlled, multicenter study (GS-US-320-1092; NCT02932150)
- Randomized phase: TAF 25 mg QD vs matched PBO for 24 weeks (completed for cohort 1 and cohort 2, group 1)
- Open-label extension phase: TAF 25 mg QD from weeks 24 to 240 (all patients, ongoing); treatment groups: TAF and PBO→TAF

Study Design

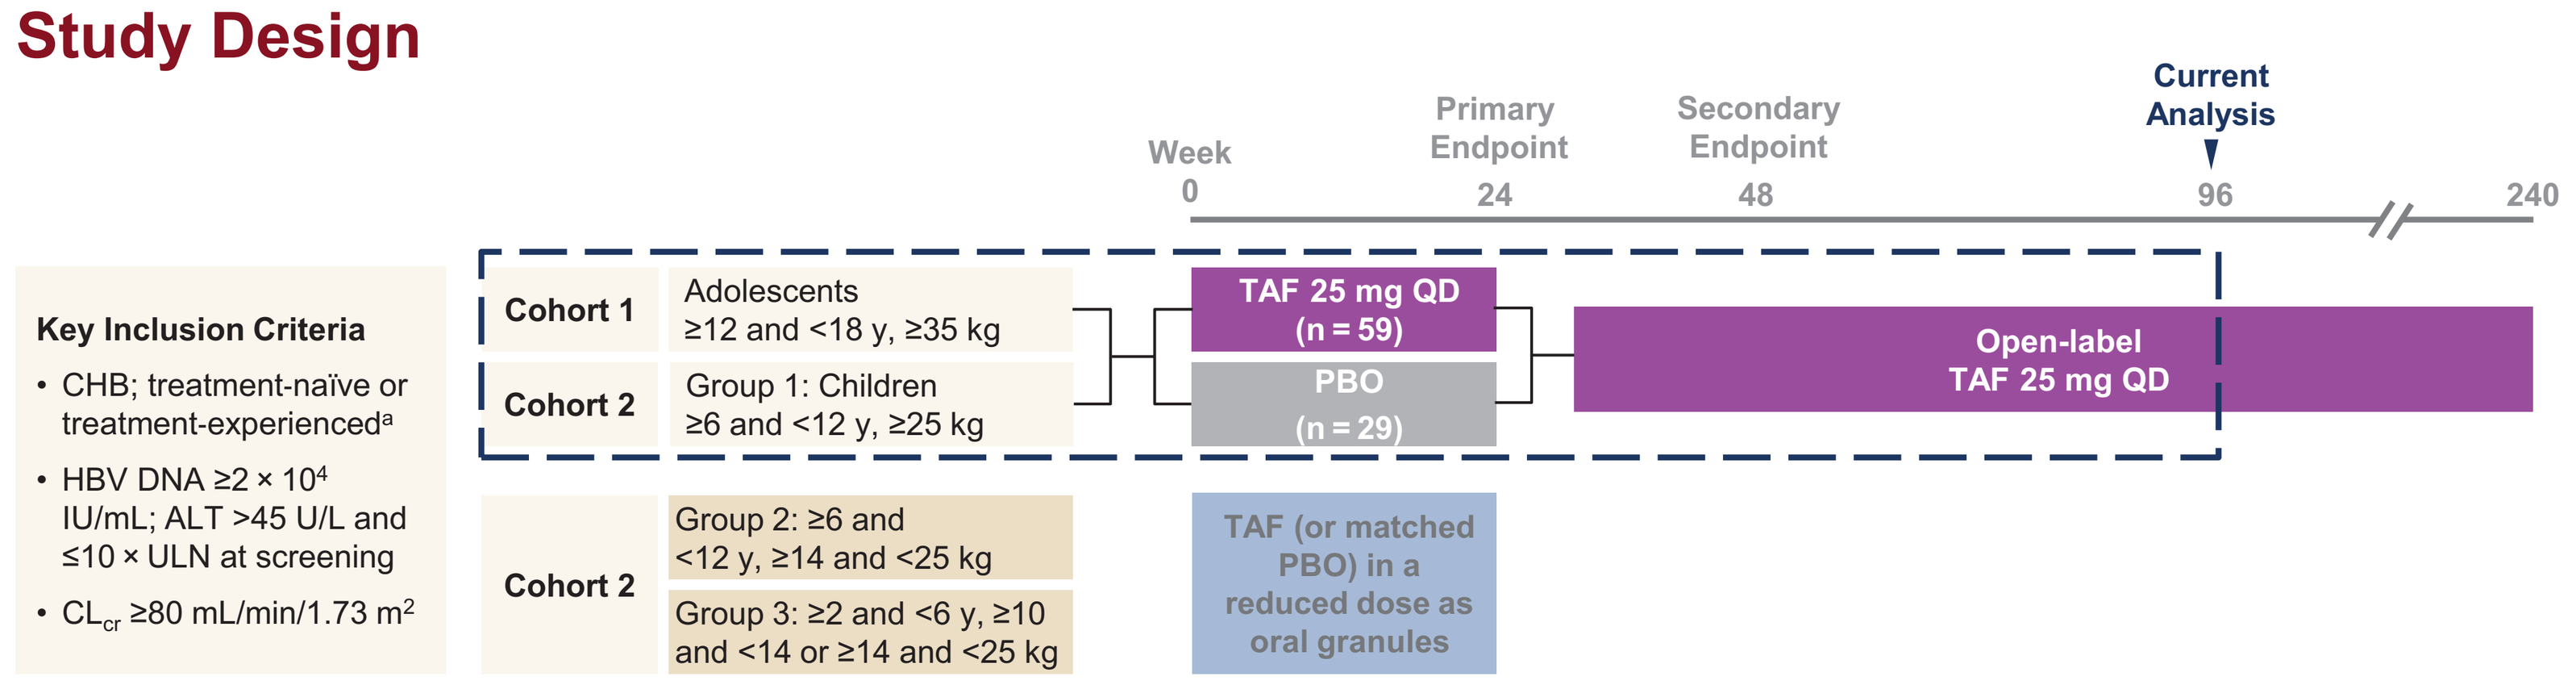
Week
0
24
48
96
240
Primary Endpoint
Secondary Endpoint
Current Analysis
Key Inclusion Criteria
• CHB; treatment-naïve or treatment-experienced[a]
• HBV DNA ≥2 × 10[4] IU/mL; ALT >45 U/L and ≤10 × ULN at screening
• CL_{cr} ≥80 mL/min/1.73 m[2]
Cohort 1
Adolescents ≥12 and <18 y, ≥35 kg
Cohort 2
Group 1: Children ≥6 and <12 y, ≥25 kg
TAF 25 mg QD (n = 59)
PBO (n = 29)
Open-label TAF 25 mg QD
Cohort 2
Group 2: ≥6 and <12 y, ≥14 and <25 kg
Group 3: ≥2 and <6 y, ≥10 and <14 or ≥14 and <25 kg
TAF (or matched PBO) in a reduced dose as oral granules

[a]Treatment-naïve: <12 weeks of treatment with any oral nucleos(t)ide analogue; treatment-experienced: ≥12 weeks of treatment with any oral nucleos(t)ide analogue.
ALT, alanine aminotransferase; CHB, chronic hepatitis B; CL_{cr}, creatinine clearance; HBV, hepatitis B virus; PBO, placebo; QD, once daily; TAF, tenofovir alafenamide; ULN, upper limit of normal; y, years.

Virology Definitions

- **Virologic breakthrough:** Hepatitis B virus (HBV) DNA ≥69 IU/mL after achieving <69 IU/mL, or a ≥1.0-$\log_{10}$ increase from nadir for 2 consecutive visits
- **Virologic blip:** HBV DNA ≥69 IU/mL after achieving <69 IU/mL or a ≥1.0-$\log_{10}$ increase from nadir for 1 visit
- **Viremia:** persistent HBV DNA ≥69 IU/mL over the course of treatment in the absence of virologic breakthrough

Conserved vs Polymorphic Amino Acid Positions

- Conserved and polymorphic positions in polymerase/reverse transcriptase (pol/RT) were previously defined in patients with CHB using treatment-naïve sequences[4]
- Conserved amino acid position
 - Only 1 amino acid observed, or
 - 2 amino acids observed, 1 of which is found in <1% of sequences
- Polymorphic amino acid position
 - 2 amino acids observed, where both are found in ≥1% of sequences, or
 - More than 2 amino acids observed, even if detected in <1% of sequences

Virology Resistance Surveillance

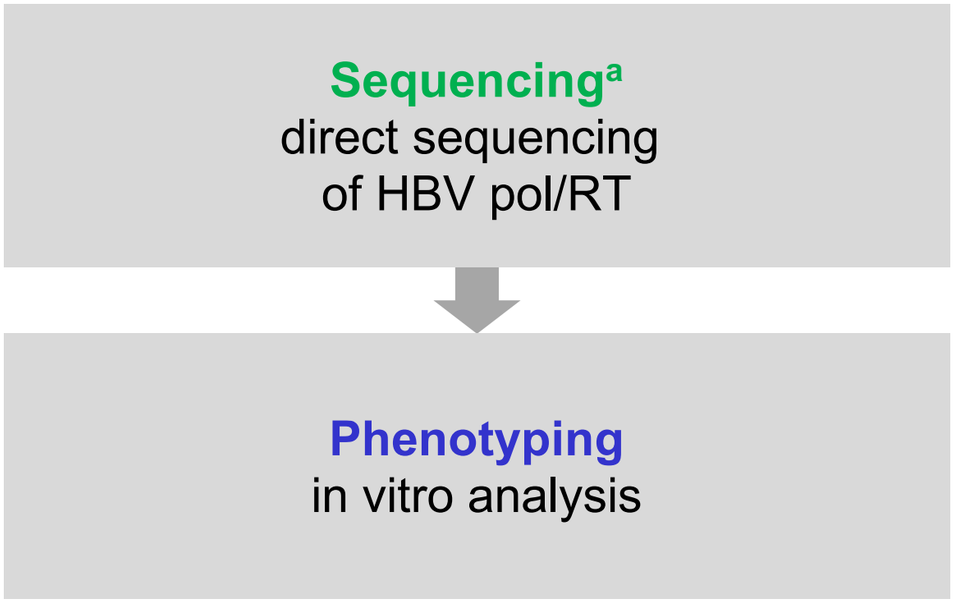
Sequencing[a]
direct sequencing of HBV pol/RT
Phenotyping
in vitro analysis

HBV pol/RT sequencing was conducted for patients:
- With HBV DNA ≥69 IU/mL at week 96, or
- At early discontinuation if HBV DNA ≥69 IU/mL

In vitro phenotyping was performed for patients with:
- Changes at a conserved site in the HBV pol/RT
- Changes at polymorphic sites if seen in >1 patient
- Virologic breakthrough with any amino acid changes in the pol/RT of HBV

[a]Limit of sequencing assay = 69 IU/mL.
HBV, hepatitis B virus; pol/RT, polymerase/reverse transcriptase.

Results

Summary of Baseline HBV Genotype Analysis

HBV Genotype	Cohort 1		Cohort 2, Group 1		Total (n = 88)
	Placebo→TAF (n = 23)	TAF (n = 47)	Placebo→TAF (n = 6)	TAF (n = 12)	
HBV genotype A	1 (4.3)	4 (8.5)	0	1 (8.3)	6 (6.8)
HBV genotype B	5 (21.7)	10 (21.3)	1 (16.7)	3 (25.0)	19 (21.6)
HBV genotype C	7 (30.4)	11 (23.4)	1 (16.7)	1 (8.3)	20 (22.7)
HBV genotype D	8 (34.8)	17 (36.2)	4 (66.7)	7 (58.3)	36 (40.9)
Mixed genotype	0	1 (2.1)	0	0	1 (1.1)
Genotype undetermined	2 (8.7)	4 (8.5)	0	0	6 (6.8)

Data are shown as n (%). HBV, hepatitis B virus; TAF, tenofovir alafenamide.

- Genotype D was the most commonly observed HBV genotype overall and represented a greater proportion in cohort 2, group 1
- Genotypes B and C were observed at similar frequencies overall and were more common in cohort 1

Patient Disposition for Resistance Testing at Week 96

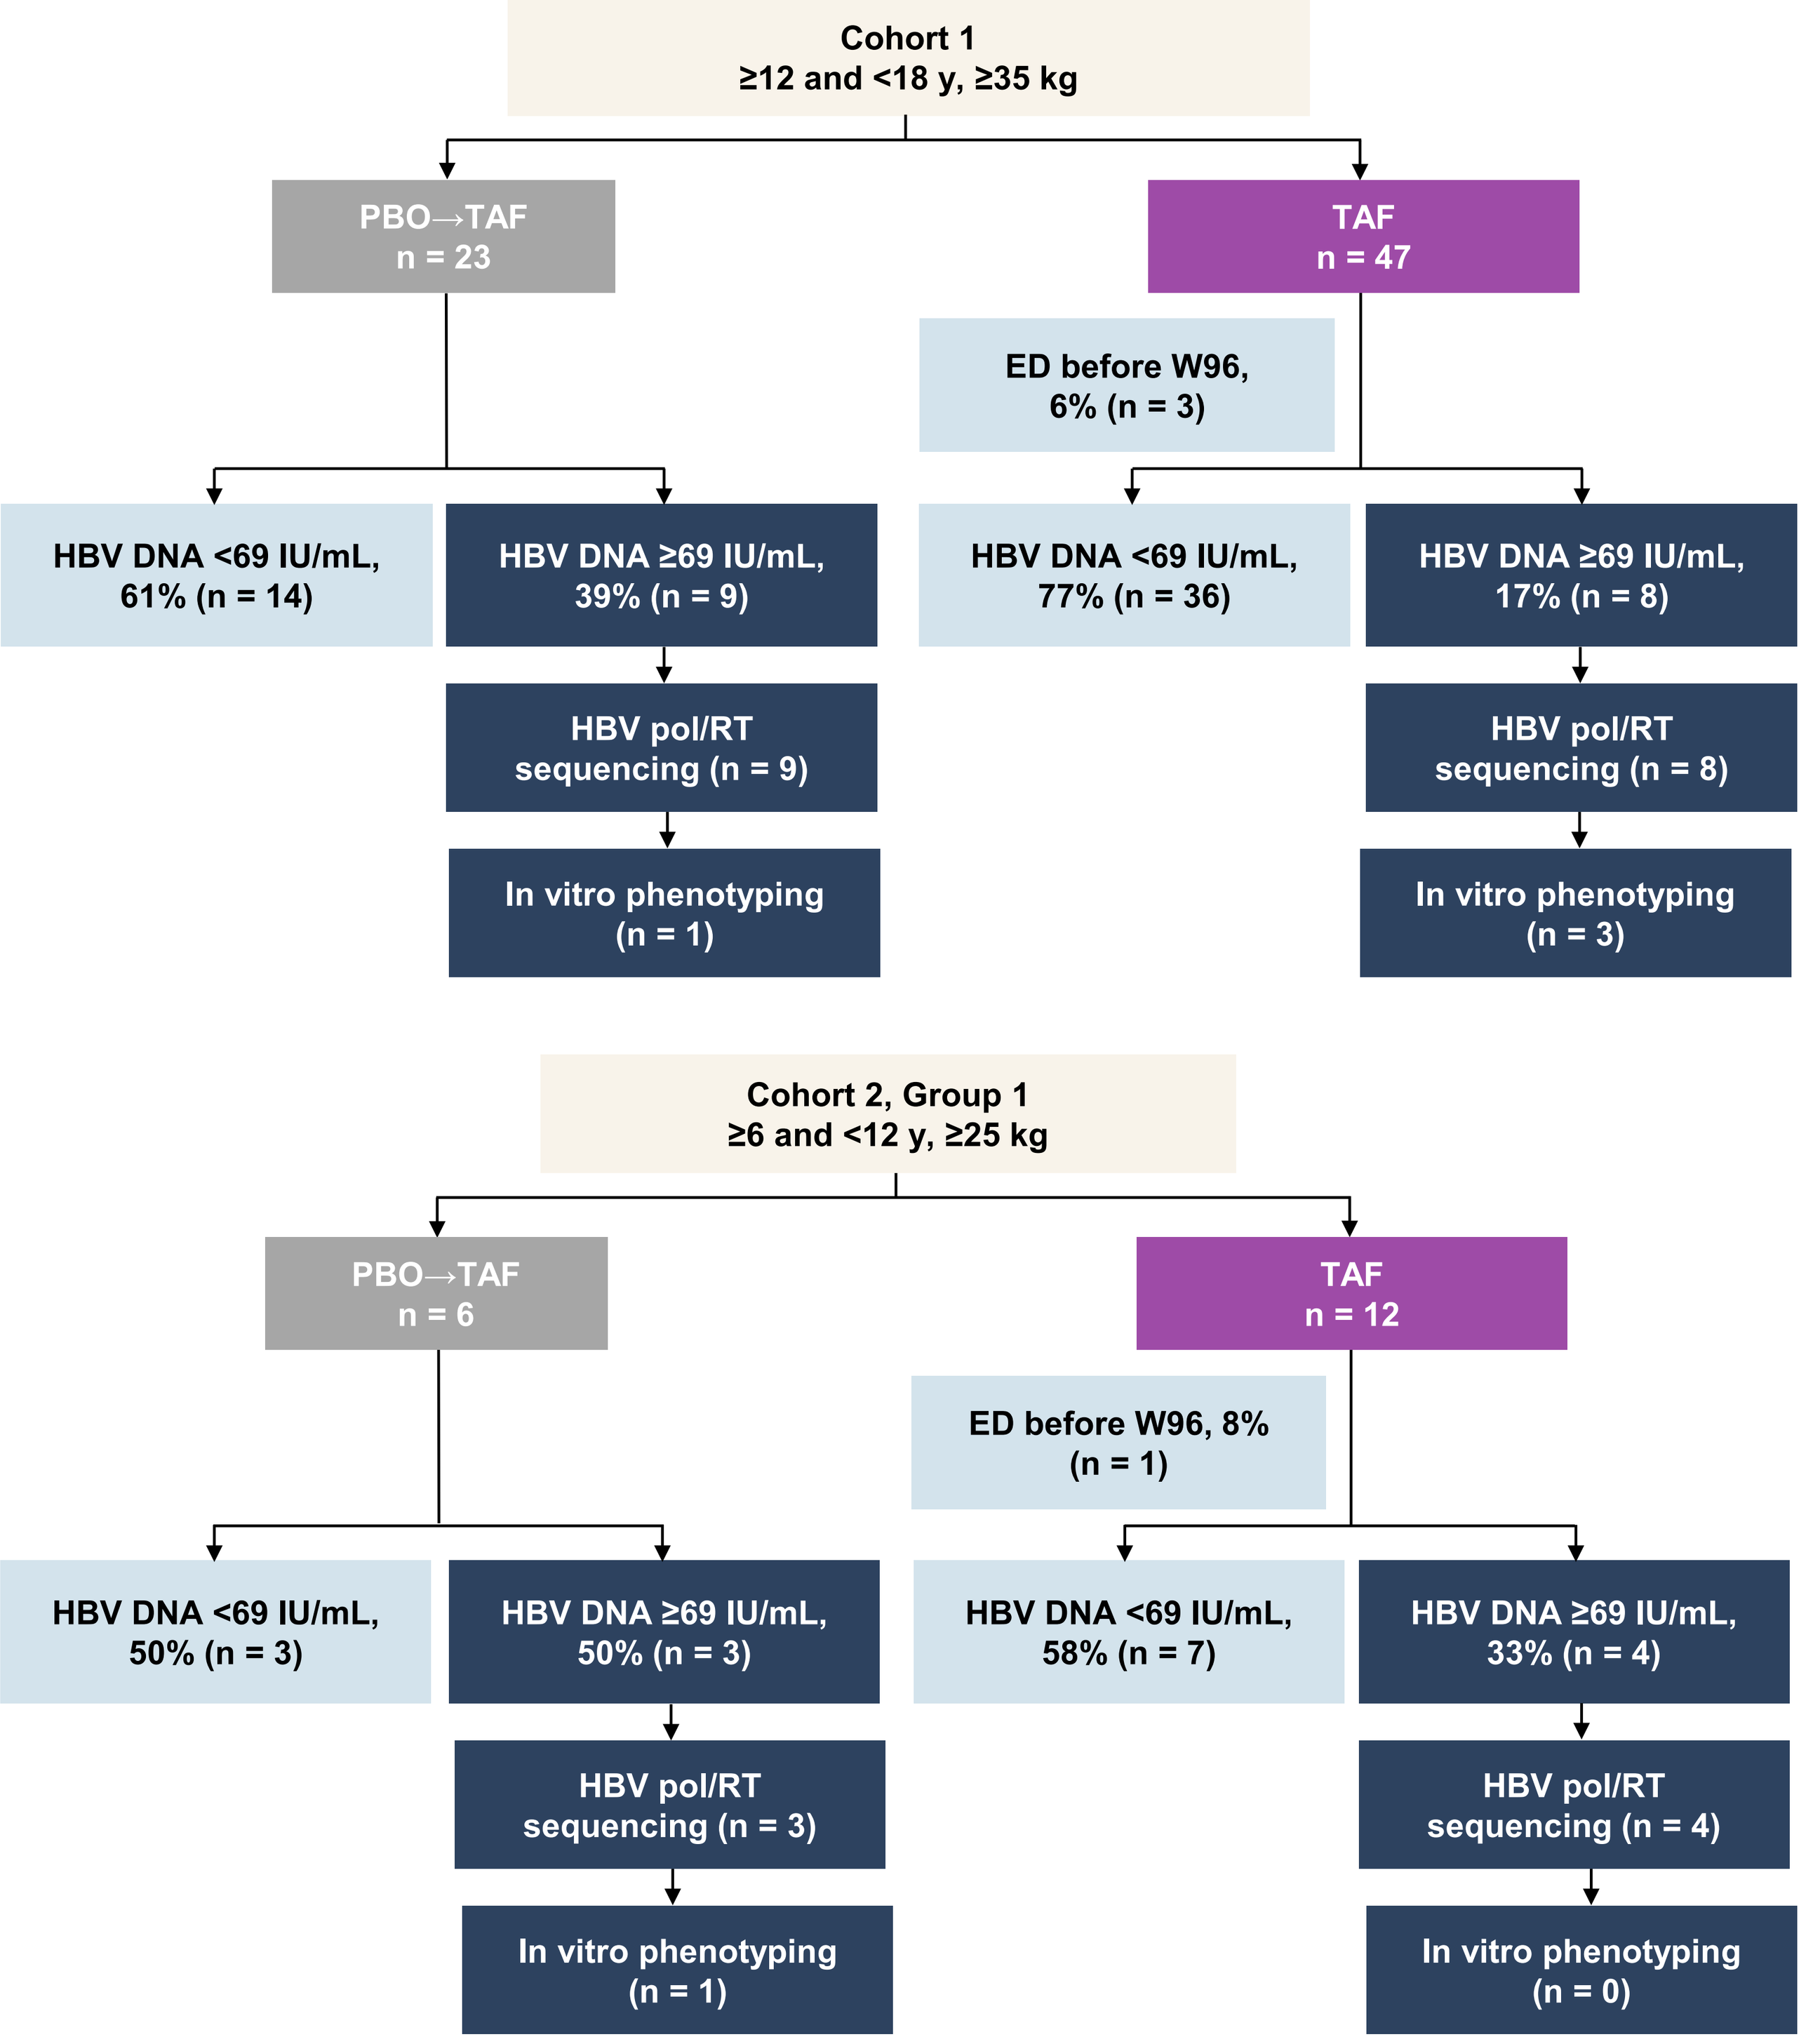
Cohort 1
≥12 and <18 y, ≥35 kg
PBO→TAF
n = 23
TAF
n = 47
ED before W96, 6% (n = 3)
HBV DNA <69 IU/mL, 61% (n = 14)
HBV DNA ≥69 IU/mL, 39% (n = 9)
HBV DNA <69 IU/mL, 77% (n = 36)
HBV DNA ≥69 IU/mL, 17% (n = 8)
HBV pol/RT sequencing (n = 9)
HBV pol/RT sequencing (n = 8)
In vitro phenotyping (n = 1)
In vitro phenotyping (n = 3)
Cohort 2, Group 1
≥6 and <12 y, ≥25 kg
PBO→TAF
n = 6
TAF
n = 12
ED before W96, 8% (n = 1)
HBV DNA <69 IU/mL, 50% (n = 3)
HBV DNA ≥69 IU/mL, 50% (n = 3)
HBV DNA <69 IU/mL, 58% (n = 7)
HBV DNA ≥69 IU/mL, 33% (n = 4)
HBV pol/RT sequencing (n = 3)
HBV pol/RT sequencing (n = 4)
In vitro phenotyping (n = 1)
In vitro phenotyping (n = 0)

ED, early discontinuation; HBV, hepatitis B virus; pol/RT, polymerase/reverse transcriptase; PBO, placebo; TAF, tenofovir alafenamide; W, week; y, years.

Reasons for Sequence Analysis at Week 96, n (%)

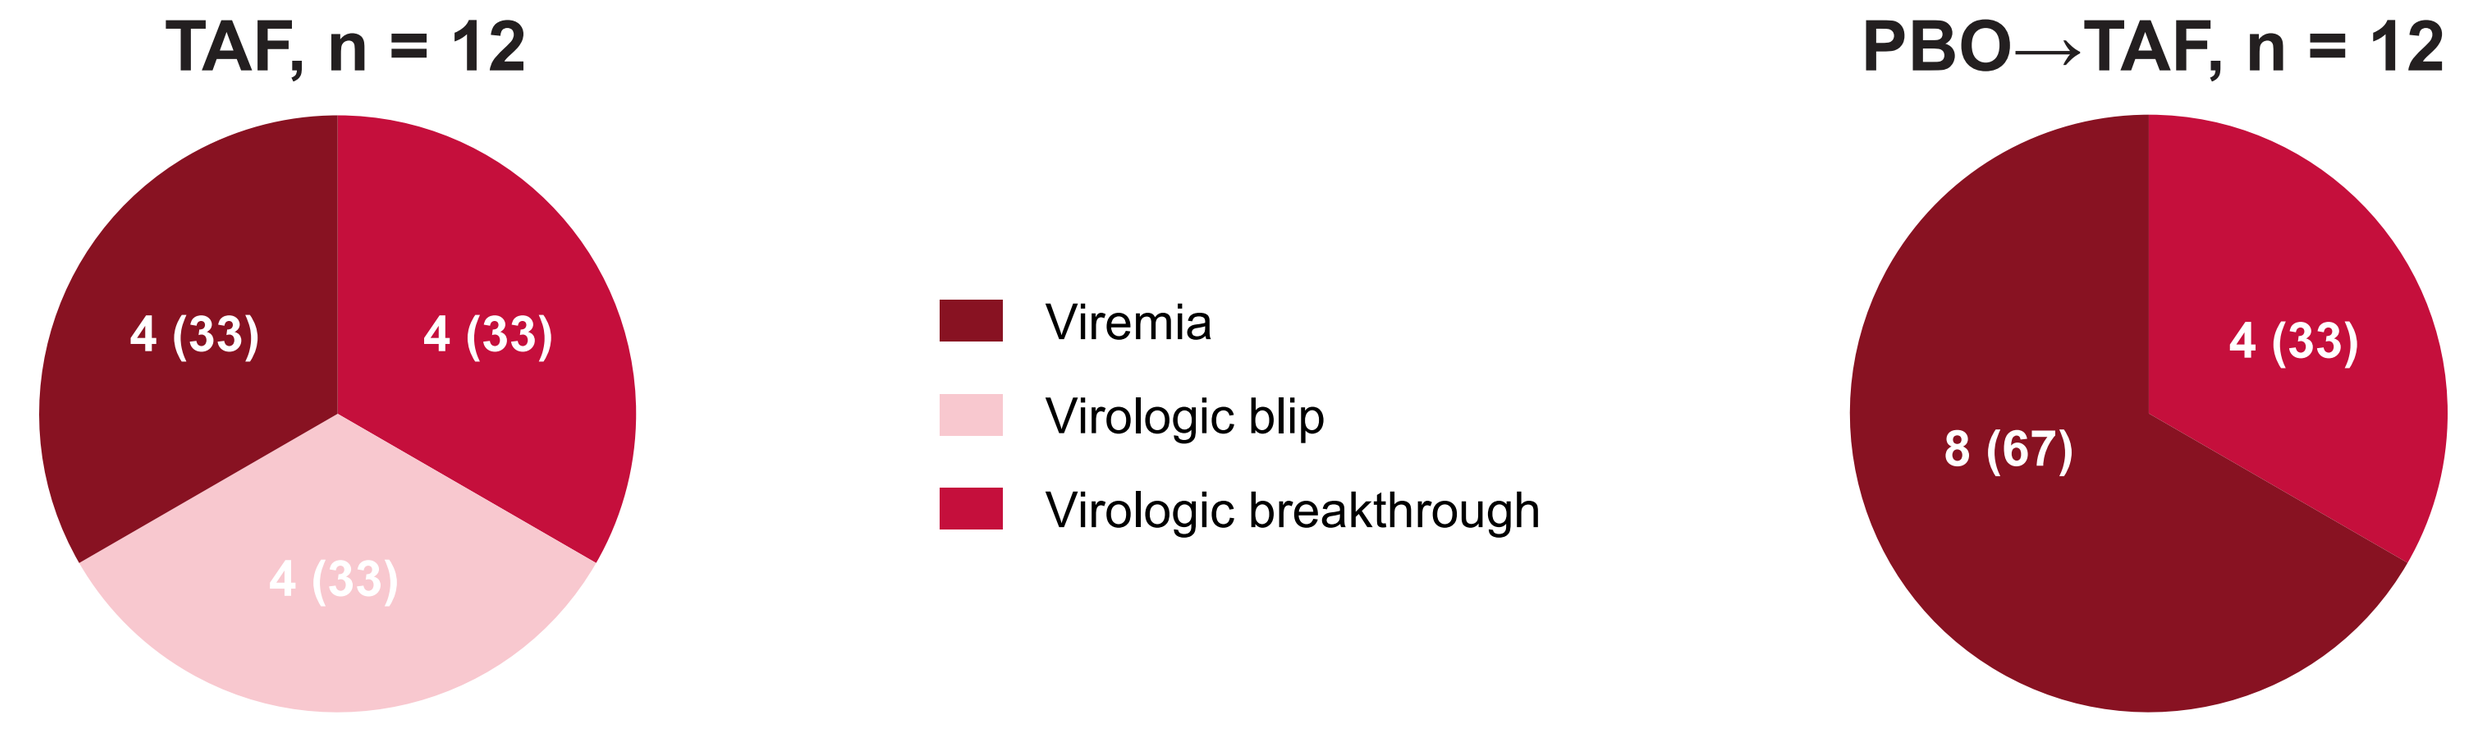
TAF, n = 12
4 (33)
4 (33)
4 (33)
Viremia
Virologic blip
Virologic breakthrough
PBO→TAF, n = 12
8 (67)
4 (33)

HBV, hepatitis B virus; PBO, placebo; pol/RT, polymerase/reverse transcriptase; TAF, tenofovir alafenamide.

- Overall, 50% of patients qualified for sequence analysis due to persistent viremia
- Similar proportions of patients (33%) qualified for virologic breakthrough

Sequence Changes in pol/RT in Patients Who Qualified for Sequence Analysis at Week 96, n (%)

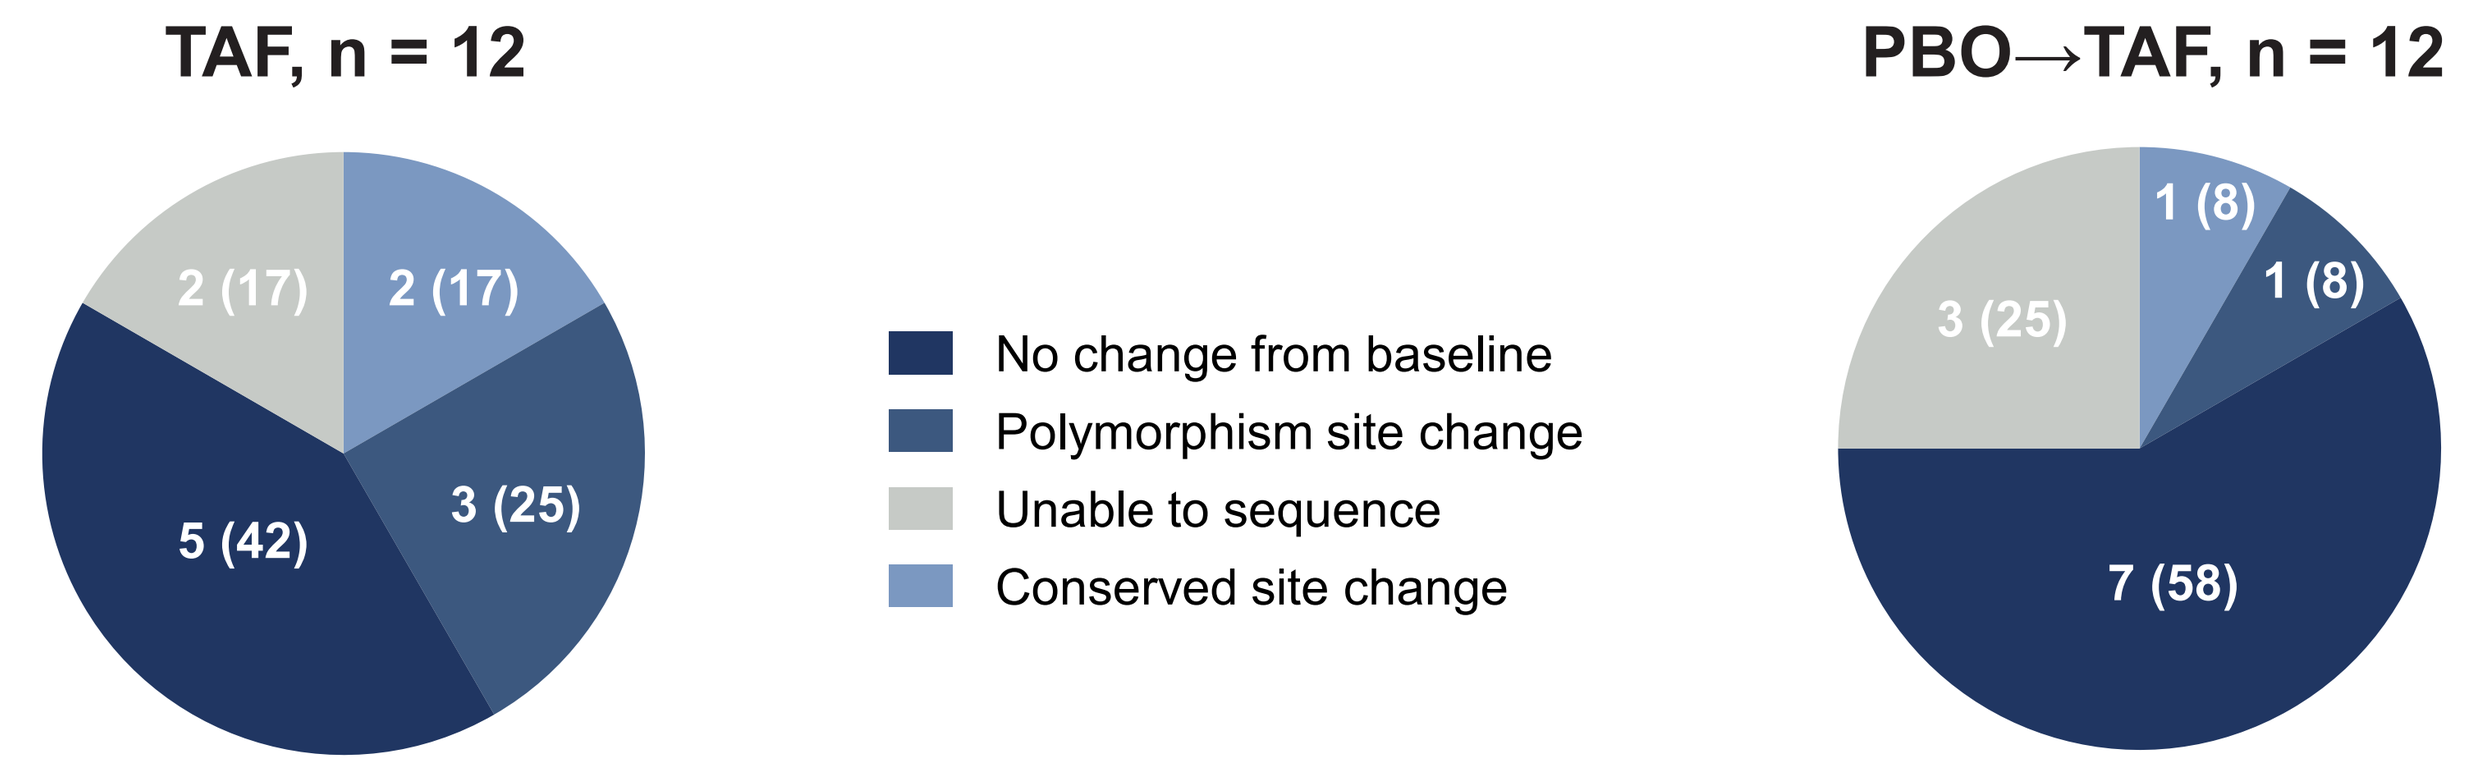
TAF, n = 12
2 (17)
2 (17)
3 (25)
5 (42)
No change from baseline
Polymorphism site change
Unable to sequence
Conserved site change
PBO→TAF, n = 12
1 (8)
1 (8)
3 (25)
7 (58)

PBO, placebo; pol/RT, polymerase/reverse transcriptase; TAF, tenofovir alafenamide.

- The majority of patients (17 of 24, 71%) had no sequence change from baseline or were unable to be sequenced
- Only 3 (12.5%) patients had conserved site changes
- Overall, 5 patients qualified for phenotypic evaluation at week 96 due to virologic breakthrough with any amino acid change (n = 2) or conserved site change (n = 3)

Phenotypic Evaluation of Qualified Patients With Available Data at Week 96[a]

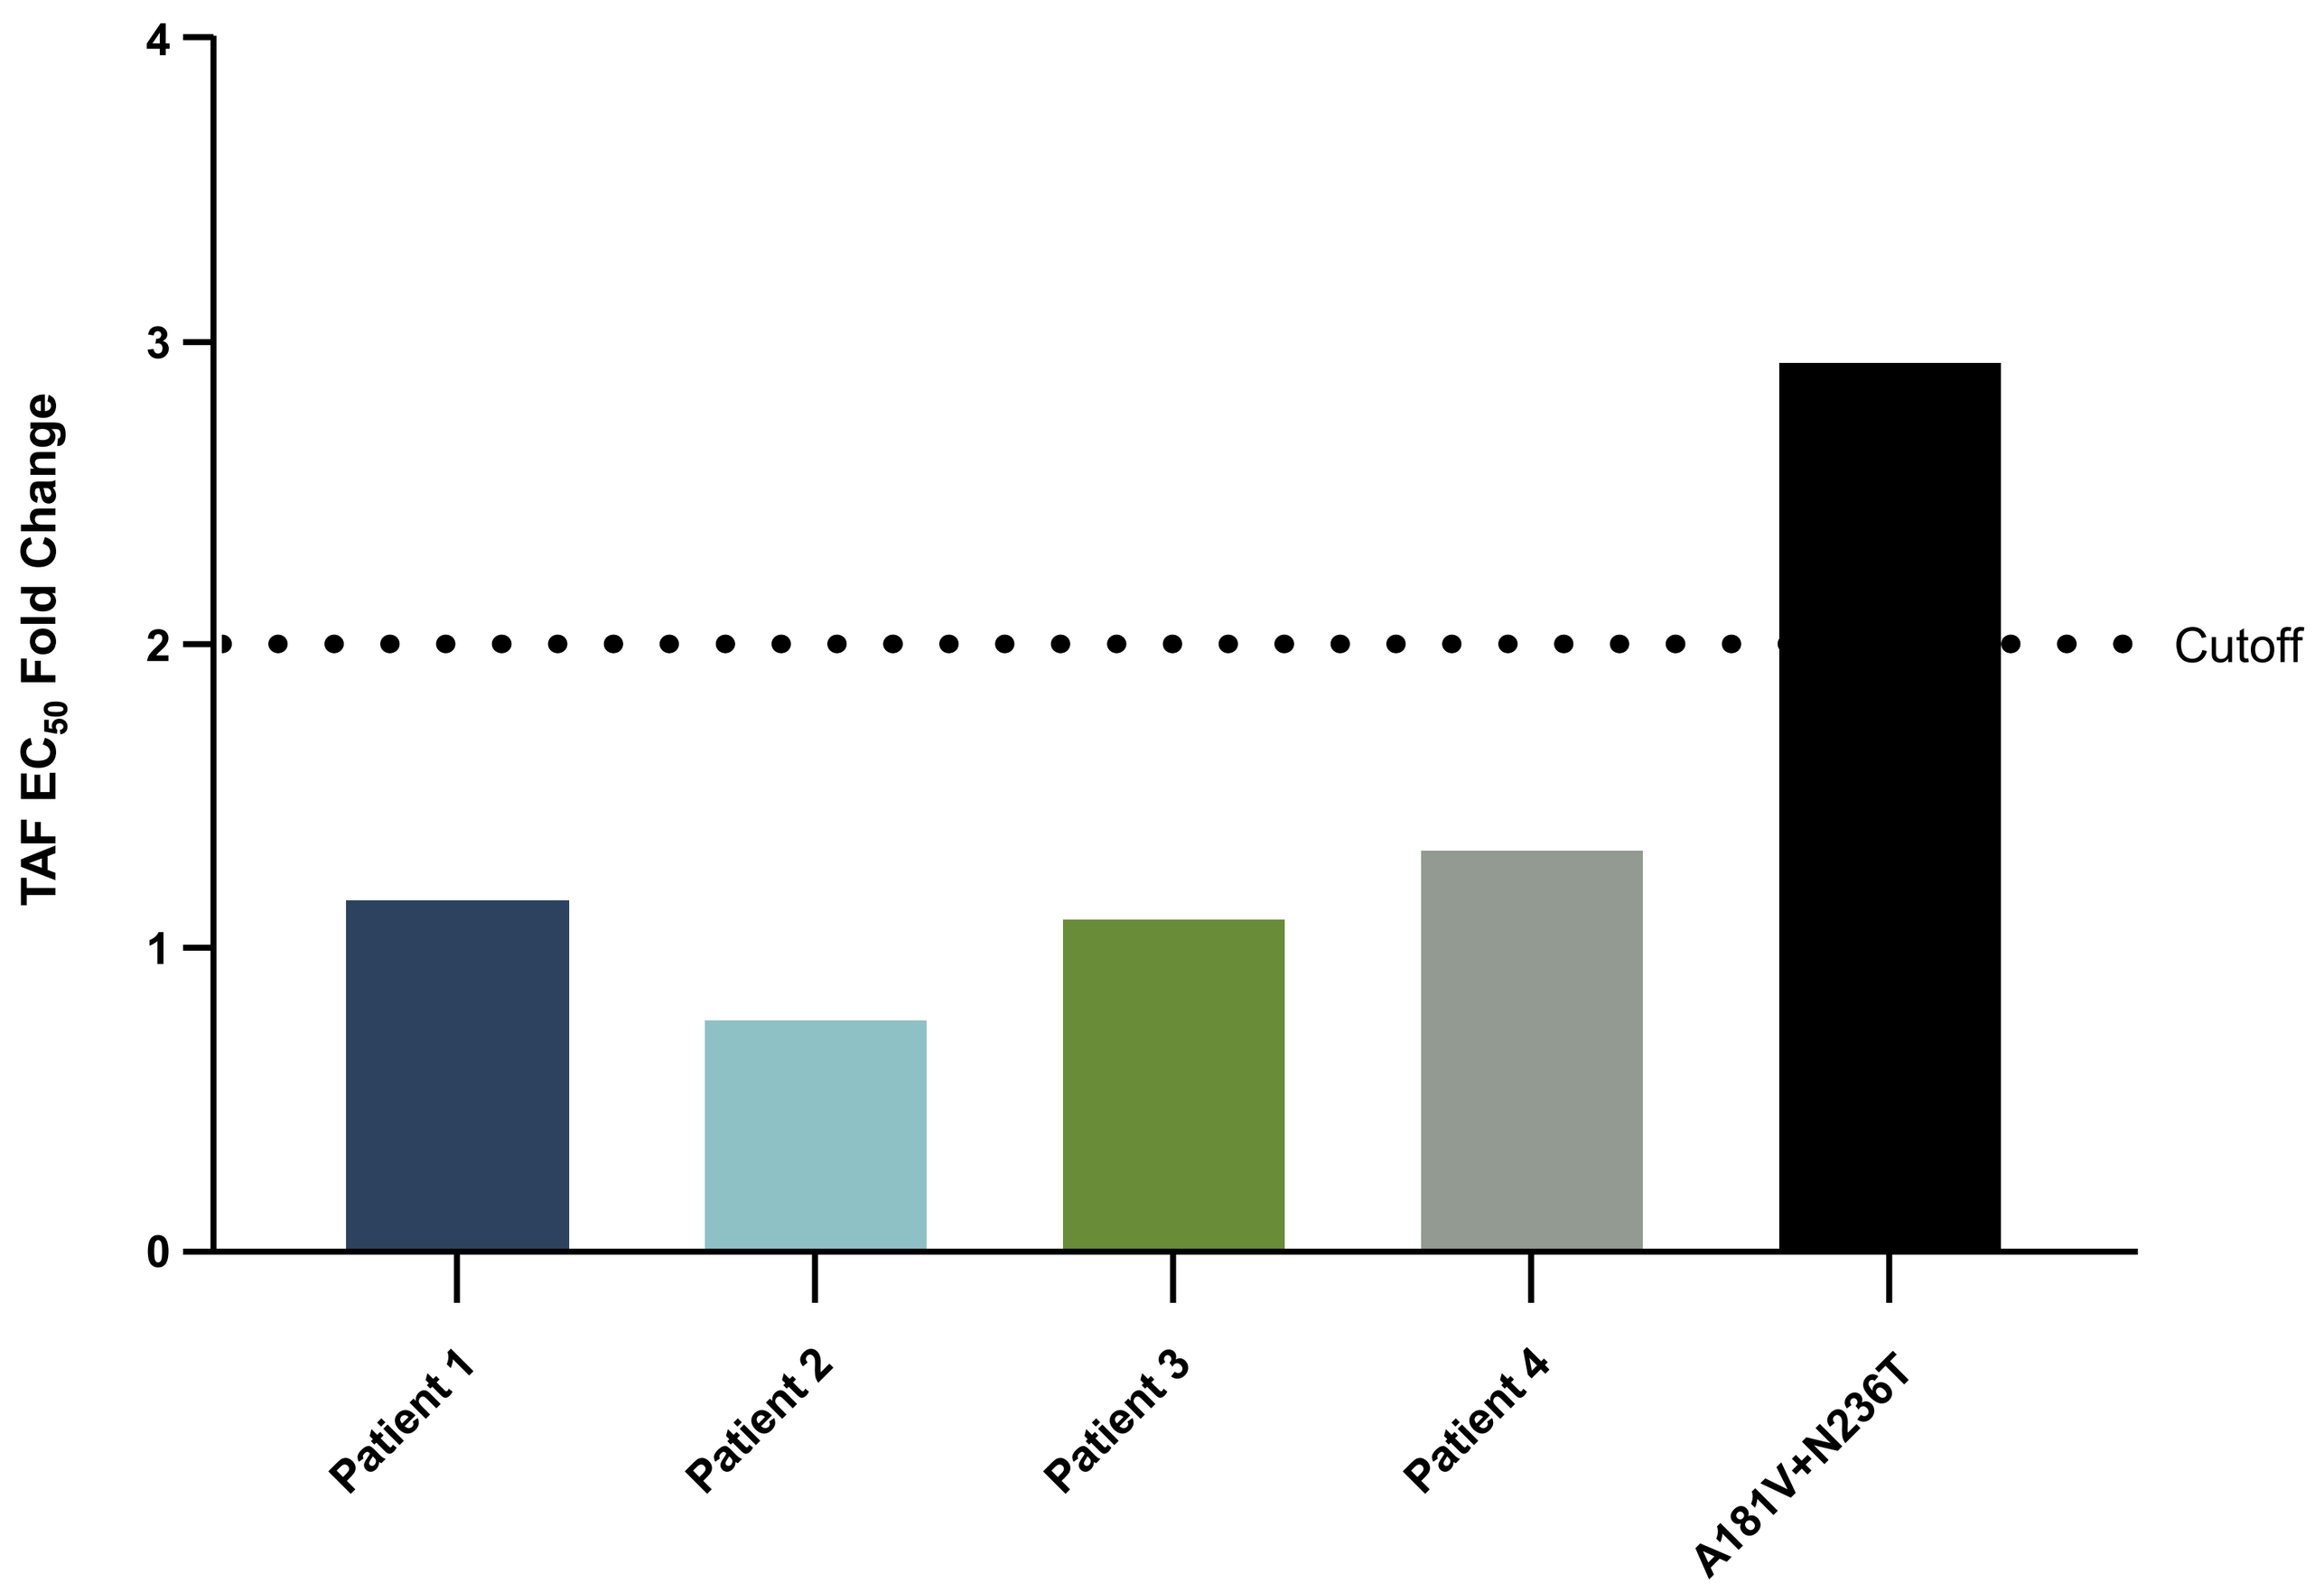
TAF EC_{50} Fold Change
0
1
2
3
4
Cutoff
Patient 1
Patient 2
Patient 3
Patient 4
A181V+N236T

[a]Phenotypic analysis was not performed for patient #5 due to sample unavailability. A181V+N236T is the assay control with low-level resistance to TAF in vitro.
EC_{50}, half-maximal effective concentration; TAF, tenofovir alafenamide.

- No patients demonstrated a >2-fold change in half-maximal effective concentration (EC_{50}) compared with baseline; therefore, no resistance to TAF was observed

AASLD: The Liver Meeting; November 10–14, 2023; Boston, MA, USA